Tuesday-49

Using Armored RNA®/DNA as Reliable Molecular Controls in Infectious Disease Testing and Beyond

Deepa Eveleigh, Maura Lloyd, and Frank Hui

Asuragen, Inc., Austin, TX

Summary

- Researchers and clinicians are always looking for controls that perform more reliably in their molecular assays.
- Asuragen has adapted its Armored technology for the use in RNA and DNA applications beyond traditional use as controls to calibrate and assess viral or cellular assay performance and sample extraction efficiency.
- We demonstrate utility of varying methods of quantifying Armored RNA and DNA upfront of more complex molecular assays to fit with our partners' needs.

Background

Recent publications have emerged related to the variability seen between the methods used to quantify reference standards[1,2]. Asuragen is working towards being able to provide a gold standard assay for quantitation of calibrators and controls. There is a clear need for robust molecular standards and process controls that can be safely used to assess the efficiency, sensitivity, and specificity of currently available and newly developed viral and cellular molecular assays. Armored technology is a system for producing robust, stable, ribonuclease-resistant nucleic acid controls and standards by assembling specific sequences and viral coat proteins into pseudo-viral particles[3]. Synthetic pseudo-biological controls provide a commutable and safe alternative to sharing positive patient samples to monitor lab-to-lab and assay-to-assay variability. Our controls are currently quantitative as calibrated against NIST-traceable standards. Production controls ensure very low lot-to-lot variability, lending to increased confidence in the stability of the process and the controls themselves. However, advancements in molecular testing including quantitative polymerase chain reaction (qPCR), droplet digital PCR (ddPCR), and next-generation sequencing (NGS) provide alternate tools that many laboratories use daily for accurate quantitation of standards and controls.

In an effort to first, further expand the accuracy and robustness of quantity assignment of the assays and second, expand the technological application of the controls, we tested different quantitation methods as comparators to standard enzymatic phosphate quantitation across multiple platforms to assess whether the Armored controls were able to produce consistent results. Here we show the value of expanding quantitative methods for RNAs for use in RT-qPCR and DNAs for use in advanced next generation sequencing technology applications.

Methods

Armored RNA Quant® (ARQ) and Armored DNA Quant (ADQ) are produced by generating target specific in vitro transcripts or PCR-amplified DNA and combining with MS2 phage capsid dimer protein particles which encapsulate the target nucleic acid and protect it from degradation (Figure 1). The resulting product is stable in plasma for up to one year at 4 degrees[4]. Asuragen's Internal Process Control (IPC) was used as the primary target for this study and used to generate both Armored RNA® and DNA product. It is a universal exogenous internal control which encapsulates a non-coding, scrambled sequence that is non-homologous to known sequences in the public domain, allowing users to design suitable primers that are distinct from own assay.

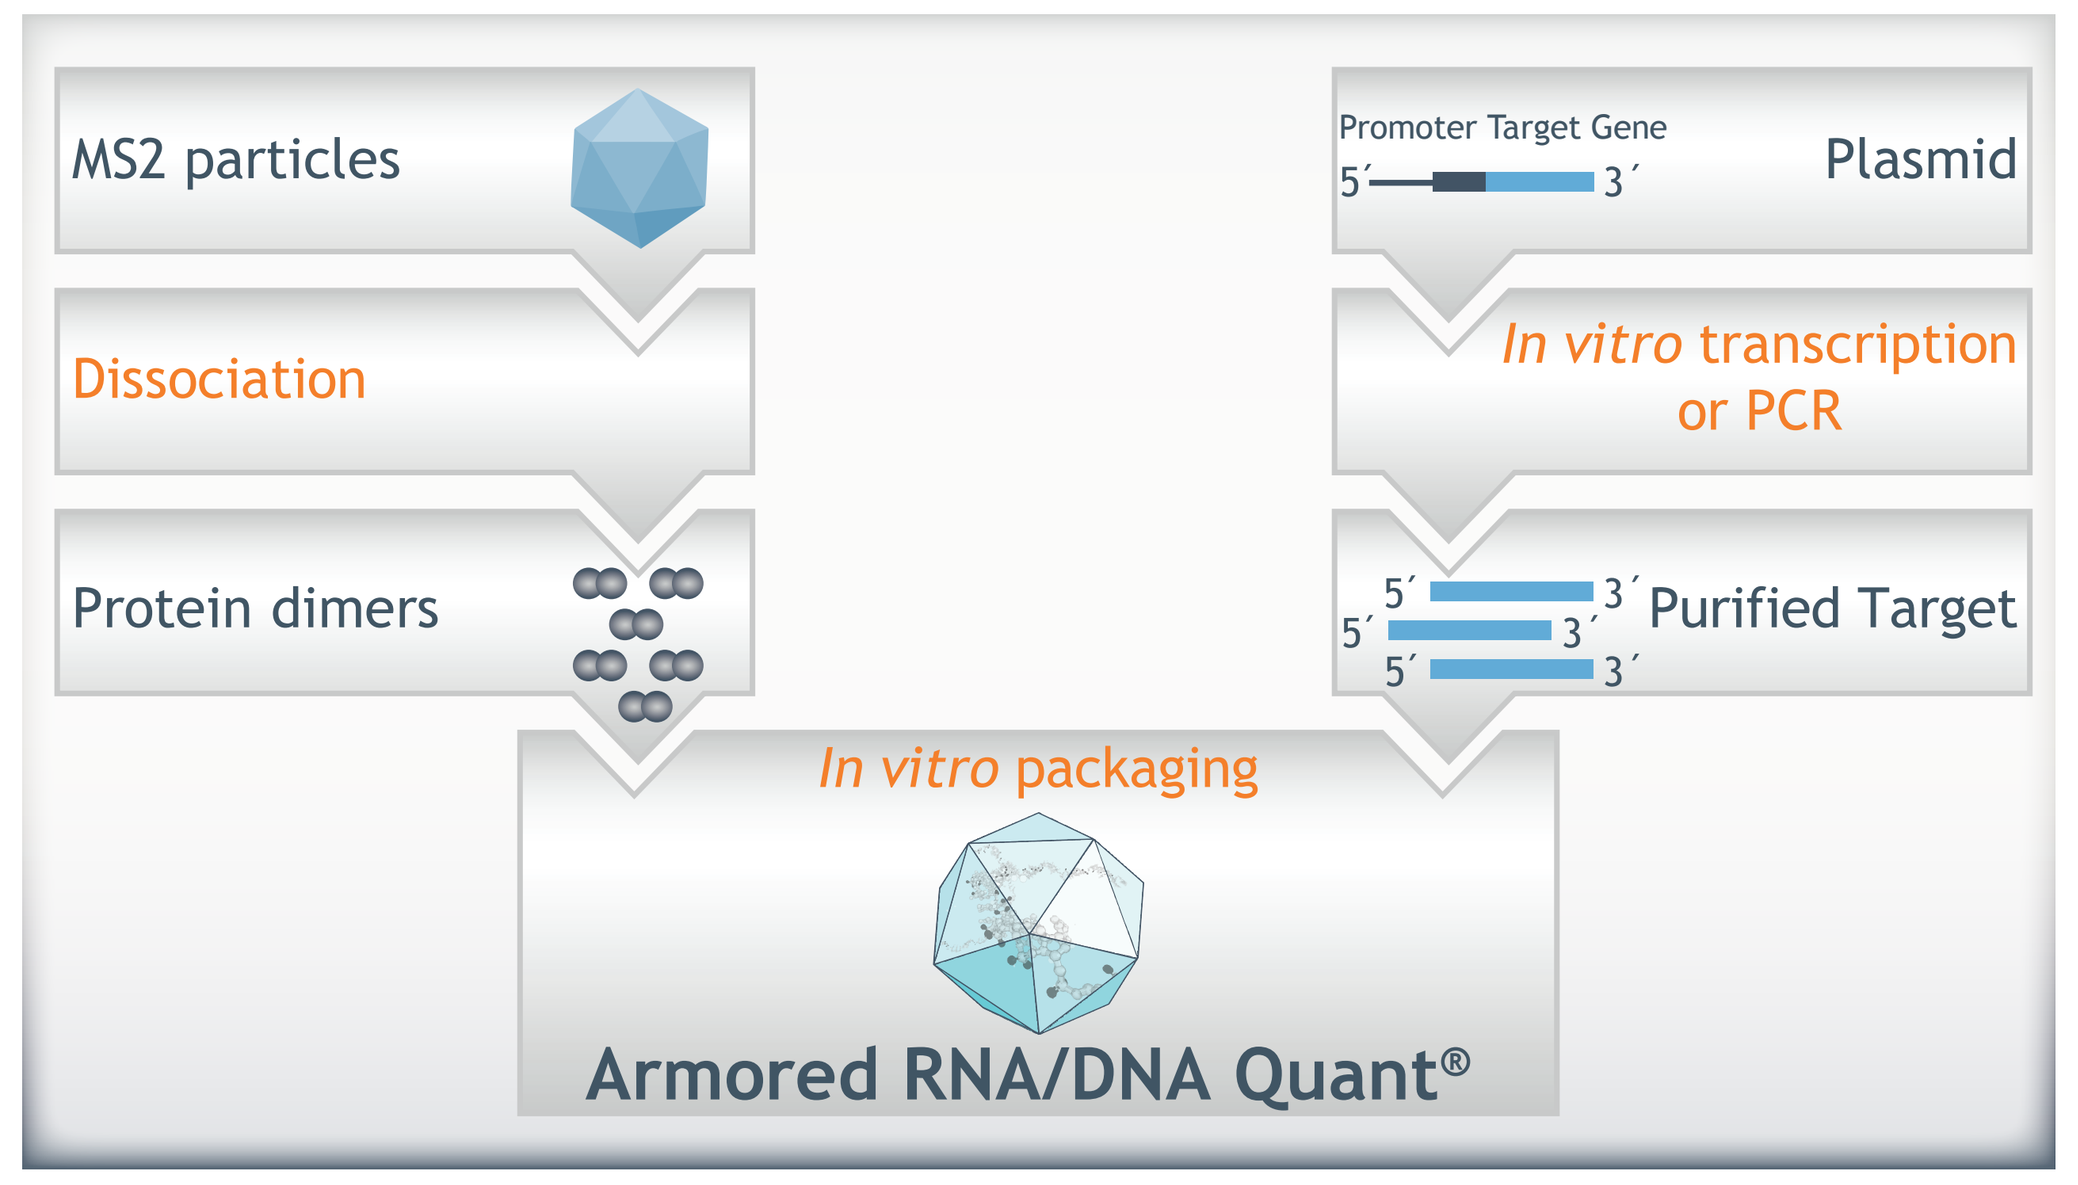
MS2 particles
Dissociation
Protein dimers
Promoter Target Gene
5′ 3′
Plasmid
In vitro transcription or PCR
5′ 3′
Purified Target
In vitro packaging
Armored RNA/DNA Quant®

Figure 1. Schematic of Armored RNA and Armored DNA Quant Production.

All test samples were heat-lysed prior to quantitation or manipulation. To quantify the Armored RNA, our validated phosphate detection method was used to measure the number of phosphate molecules released from the isolated material compared to a NIST-traceable standard. For RT-qPCR detection of the IPC, Applied Biosystems 7500Fast Dx instrumentation was used with TaqMan chemistry. Heat-lysed ARQ was serially diluted and run alongside an IVT standard curve that was quantified using spectrophotometry. The Armored DNA was quantified using Bioanalysis on a High Sensitivity DNA chip from Agilent. ddPCR was performed using BioRad's QX200 Auto DG and Droplet Reader System to obtain absolute copies. qPCR was carried out on a 7500Fast Dx system alongside a KAPA standard for quantitation (KAPA Biosystems). For NGS sequencing, libraries were generated using Asuragen's QuantideX library reagents and sequencing on Illumina's MiSeq System. Sizing of libraries and PCR products was determined using the Agilent Bioanalyzer.

Presented at CVS 2019

Results

The IPC Armored RNA was first quantified by phosphate assay and measured to be 2E9 copies/mL at the highest dilution used. IPC IVT and IPC ARQ heat-lysed RNA were titrated in one log increments from the same starting concentration (2E9 copies/mL) based on the phosphate assay and Ct measurement taken by RT-qPCR as shown in Figure 2. Phosphate vs RT-qPCR measurement evaluation as compared to spectrophotometry for both IVT and ARQ is summarized in Table 1. Although formulated to equivalent levels, there is a shift in the RT-qPCR measured concentrations between the Armored RNA and IVT of about 33% on average. This magnitude of a shift is not significant for an application such as qPCR. For the IPC Armored DNA, concentration was first assessed by Bioanalysis (Figure 3). Serial dilutions of lysed DNA were also submitted for ddPCR and qPCR testing with results showing expected linearity across all dilutions (Figures 4a and 4b). Back-calculation of serial dilutions was performed for each dilution series point to obtain an average concentration of what the stock value was calculated to be for the IPC DNA (Table 1). Based on the values obtained, observed differences were 20-30% of expected based on analytical method. The IPC DNA was also able to be successfully run on the Illumina MiSeq System. The sequencing reads were of sufficient quantity and quality to be able to extract and align to the IPC target amplicon reference sequence with high fidelity (data not shown).

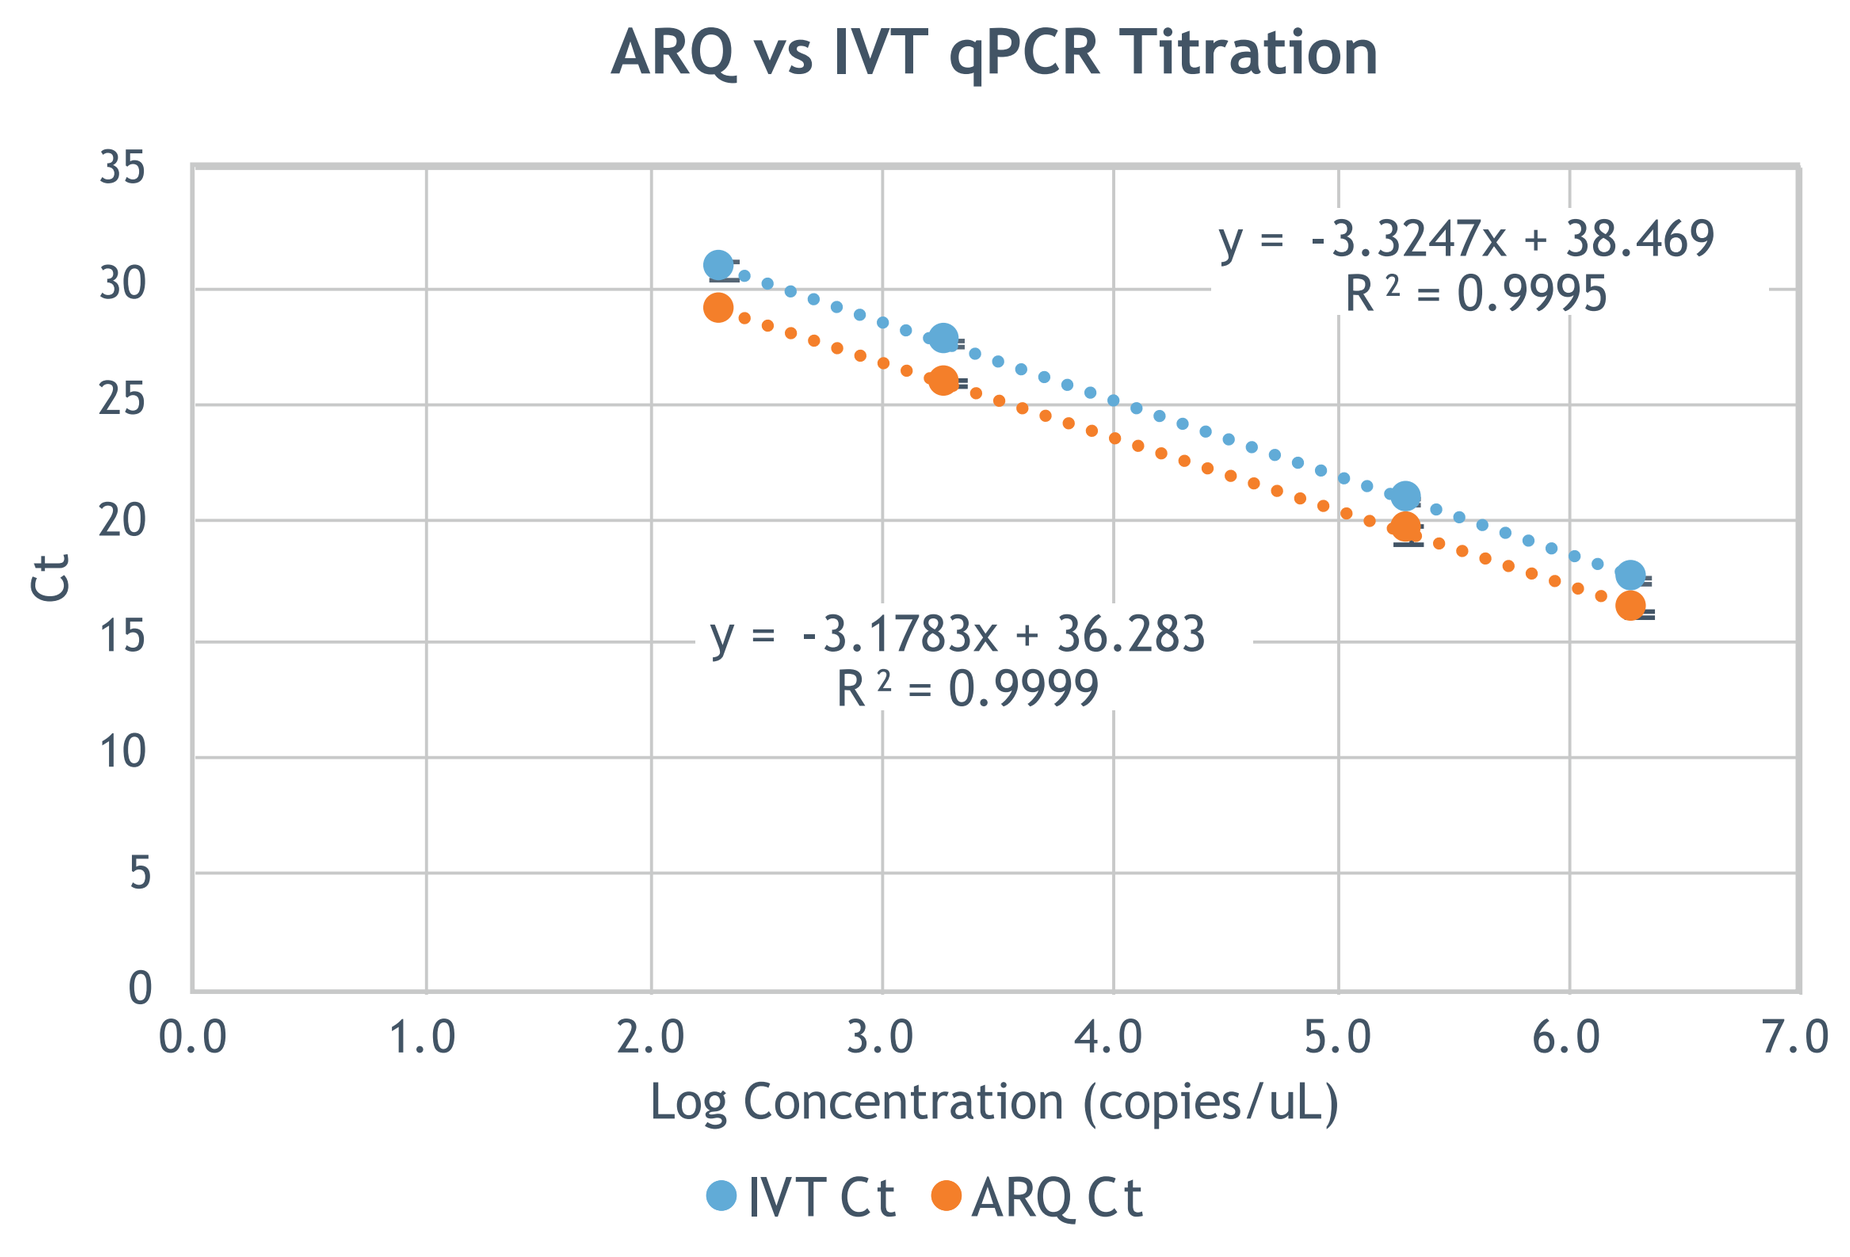
ARQ vs IVT qPCR Titration
y = -3.3247x + 38.469
R² = 0.9995
y = -3.1783x + 36.283
R² = 0.9999
Ct
0
5
10
15
20
25
30
35
0.0
1.0
2.0
3.0
4.0
5.0
6.0
7.0
Log Concentration (copies/uL)
IVT Ct
ARQ Ct

Figure 2. IPC IVT RNA Titration Series Cts vs ARQ Heat-Lysed RNA Titration Series Cts. A difference of approximately 33% is seen when back calculating copies/mL of stock for each type of RNA.

Table 1. Results in Copies/mL of Various Quantitation Methods Back-Calculated to Stock.

Analyte	Quantitation Method	Average cp/mL
ARQ	Phosphate assay	2.00E+09
ARQ	RT-qPCR	6.01E+09
ADQ	Bioanalyzer	2.72E+10
ADQ	Droplet Digital	6.01E+09
ADQ	qPCR	7.98E+09
IVT	Phosphate assay	9.23E+15
IVT	Spectrophotometer	8.70E+15

ARQ = Armored RNA Quant and ADQ = Armored DNA Quant generated from Asuragen's IPC target.

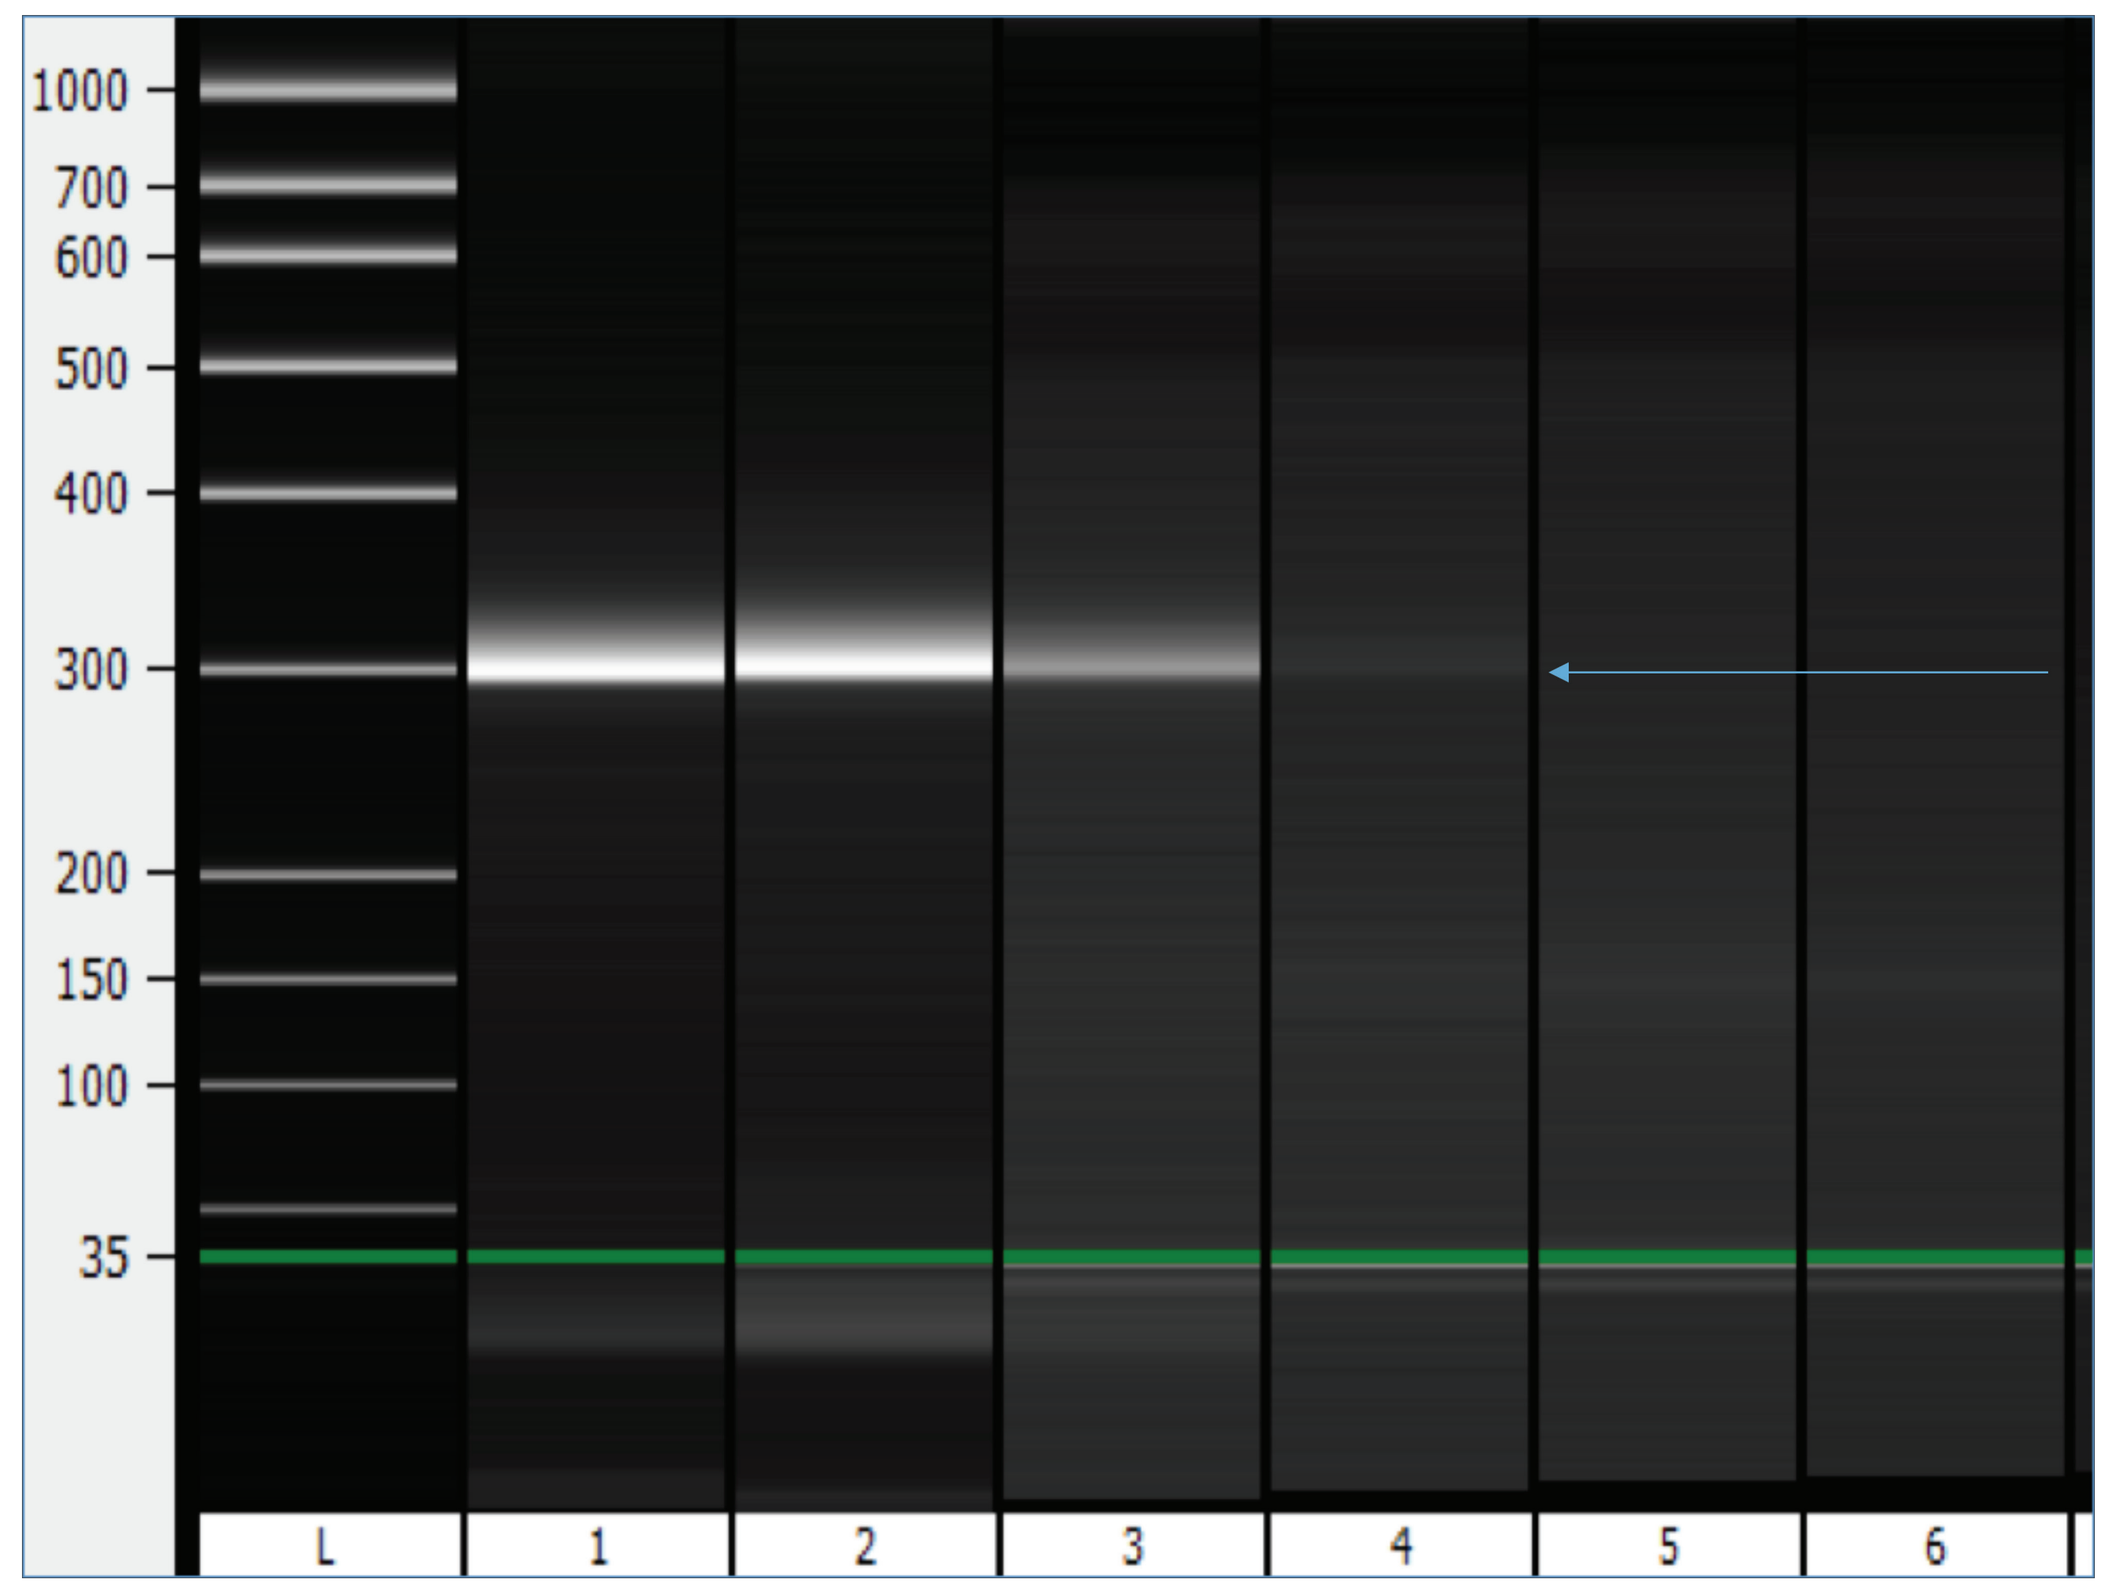
1000
700
600
500
400
300
200
150
100
35
L
1
2
3
4
5
6

Figure 3. Bioanalysis Profile of Serially Diluted IPC Armored DNA Quant. L=ladder, 1=heat-lysed stock DNA, 2=10-1 dilution, 3=10-2 dilution, 4=10-3 dilution, 5=10-4 dilution, 6=10-5 dilution. A distinct band (arrow) is seen down to the 10-3 dilution.

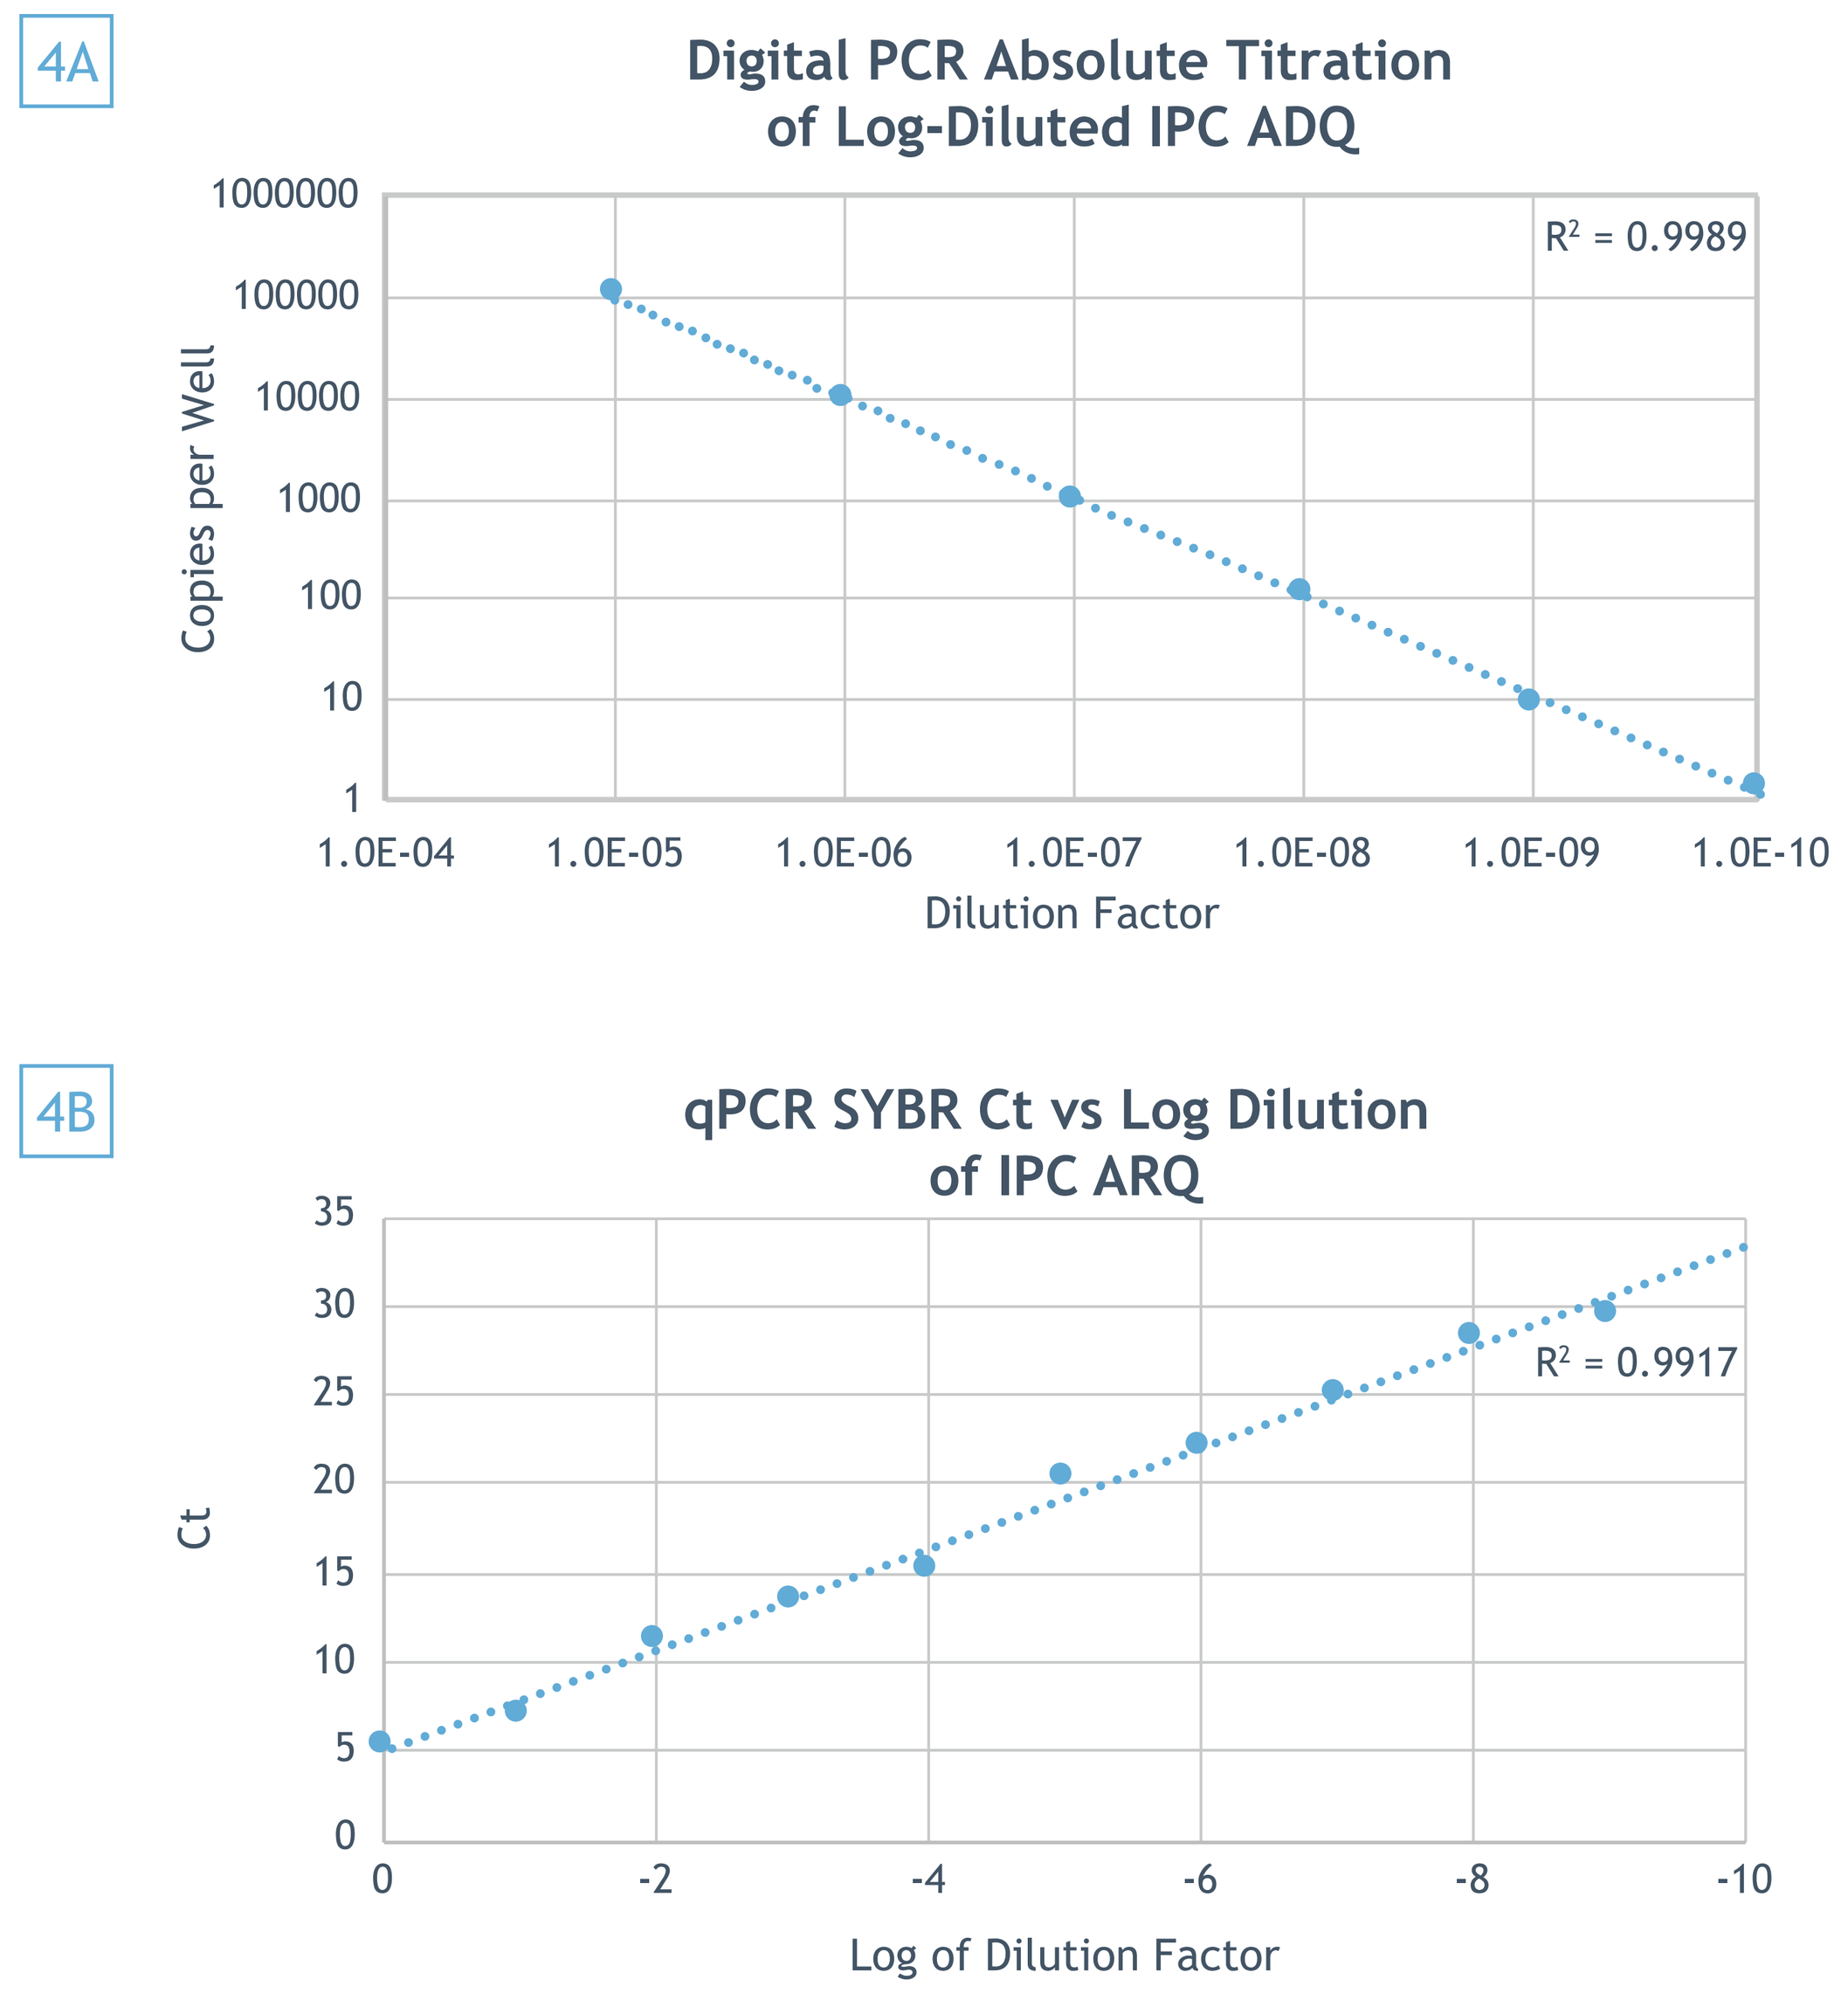
4A
Digital PCR Absolute Titration of Log-Diluted IPC ADQ
R² = 0.9989
Copies per Well
1000000
100000
10000
1000
100
10
1
1.0E-04
1.0E-05
1.0E-06
1.0E-07
1.0E-08
1.0E-09
1.0E-10
Dilution Factor
4B
qPCR SYBR Ct vs Log Dilution of IPC ARQ
R² = 0.9917
Ct
35
30
25
20
15
10
5
0
0
-2
-4
-6
-8
-10
Log of Dilution Factor

Figure 4. Dilution Series Measurement Shows Good Target-Specific Linearity Across Digital A) or quantitative B) PCR Assay.

Conclusions

- We have demonstrated that traditional methods for quantitation of Armored RNA and Armored DNA are enriched by using more complex molecular techniques to standardize or normalize value assignments.
- Asuragen has enhanced the technology offering by expanding customization to better align with research and diagnostic needs.
- This allows for extension of applications from traditional clinical virology studies to integration with assays used in a variety of different therapeutic areas beyond infectious disease.
- The alternative methods described here including qPCR, ddPCR, RT-qPCR and NGS are reliable and offer the flexibility for end users to select an optimal readout for their particular molecular assay needs.

References

1. Bateman, A.C., et al., Quantification of BK Virus Standards by Quantitative Real-Time PCR and Droplet Digital PCR Is Confounded by Multiple Virus Populations in the WHO BKV International Standard Clin Chem, 2017, 63(3):761-769
2. Hayden, R.T., et al., Comparison of droplet digital PCR to real-time PCR for quantitative detection of cytomegalovirus. J Clin Microbiol, 2013, 51(2):540-6
3. WalkerPeach, C.R., et al., Ribonuclease-resistant RNA controls (Armored RNA) for reverse transcription-PCR, branched DNA, and genotyping assays for hepatitis C virus. Clin Chem, 1999, 45(12):2079-85
4. Asuragen internal technical reports

Scan to download of PDF file of this poster

Asuragen®